IHC-PO-387

Presented at
International Headache Congress (IHC);
5-8 September 2019;
Dublin, Ireland.

Patient Satisfaction With Fremanezumab Treatment in Migraine: Results From a Patient Survey After Completion of a 1-year Extension Study

Richard B. Lipton,[1] Sanjay K. Gandhi,[2] Joshua M. Cohen,[2] Blaine Cloud,[3] Verena Ramirez-Campos,[2] Andrew H. Ahn,[2] Dawn C. Buse[1]

[1]Department of Neurology, Albert Einstein College of Medicine, New York, NY, USA; [2]Teva Pharmaceuticals Industries, Frazer, PA, USA; [3]Clinical SCORE, Chadds Ford, PA, USA.

CONCLUSIONS

- **Treatment satisfaction with fremanezumab was high overall and across different treatment dimensions in patients with chronic migraine (CM) and episodic migraine (EM)**
- **Patients were generally satisfied with their overall experience with fremanezumab, and patients commonly reported being satisfied with the reduced frequency of migraine attacks with fremanezumab**
- **Results of this survey may be limited by recall and participation bias; however, the sample was representative of the overall treatment population, and results were similarly distributed regardless of diagnosis (CM vs EM) or dosing regimen received (quarterly vs monthly)**

OBJECTIVES

- Fremanezumab is a fully humanized monoclonal antibody (IgG2Δa) that selectively targets calcitonin gene-related peptide (CGRP)[1]
 - Proven efficacy for the preventive treatment of migraine in adults[2,3]
- Long-term safety and efficacy of fremanezumab were evaluated in a 52-week extension study
- Patient satisfaction with fremanezumab treatment was evaluated using a retrospective, web-based questionnaire in a subpopulation of patients who completed the extension study and consented to participate in the survey

METHODS

Patients

- In the 52-week extension study, adults ≥18 years of age with CM or EM were randomized to the following:
 - Quarterly fremanezumab (675 mg)
 - Monthly fremanezumab (225 mg)
 - Some CM patients received a loading dose of 675 mg of fremanezumab in the monthly arm
- During the extension study, all patients were blinded to the treatments they were receiving
- Patients were recruited at 41 extension study sites in the United States to participate in the retrospective survey
- Patients who started another CGRP-targeted migraine treatment after the extension study were excluded from participating in the survey
- After consenting to participate, patients were directed to an online web survey

Study Assessments

- Patients completed an online patient experience survey (~20-40 minutes)
- As part of that retrospective survey, patients reported satisfaction overall and across different aspects of treatment and treatment dimensions, based on the questions shown in **Table 1**

Table 1. Survey Questions and Response Options to Evaluate Patient Satisfaction With Fremanezumab Treatment

1. How satisfied or dissatisfied were you with the ability of the clinical trial medication to prevent or treat your migraine?
2. How satisfied or dissatisfied were you with the way the clinical trial medication relieved your migraine symptoms?
3. How satisfied or dissatisfied were you with the amount of time it took the clinical trial medication to start working?
4. How easy or difficult was it to have the clinical trial medication administered in the clinical trial site (doctor's office)?
5. Taking all things into account, how satisfied or dissatisfied were you with the clinical trial medication?

Extremely dissatisfied/ difficult[a]			Neither dissatisfied nor satisfied/difficult nor easy[a]			Extremely satisfied/ easy[a]
1	2	3	4	5	6	7

6. Thinking specifically of the experimental medicine's ability to prevent migraine, with which dimension of your migraine relief were you satisfied? *Select all that apply.*

- ☐ Reducing attack frequency
- ☐ Reducing migraine pain intensity
- ☐ Reducing attack duration
- ☐ Reducing migraine-associated symptoms (like nausea, light & sound sensitivity)
- ☐ Reducing migraine-associated disability (ability to work and participate in activities)
- ☐ None of the above

[a]Responses ranging from extremely difficult to extremely easy were for question 4 only; all other questions used the responses ranging from extremely dissatisfied to extremely satisfied.

RESULTS

Patient Characteristics

- 51% (N = 302) of patients consented to take the retrospective survey
 - Completed survey 1 to 24 months after last extension study visit, n = 253
 - Excluded due to use of another CGRP-targeted migraine treatment, n = 49
- All patients received active treatment during the extension study; 134 also received fremanezumab during a prior phase 3 study
- Demographic and baseline characteristics are summarized in **Table 2**

Table 2. Demographic and Baseline Characteristics

Characteristic	Total (n = 253)	CM (n = 134)	EM (n = 119)	Quarterly (n = 131)	Monthly (n = 122)
Mean (SD) age, years	45.5 (11.6)	45.2 (11.0)	46.9 (12.3)	47.3 (10.9)	44.7 (12.2)
Mean (SD) age at migraine diagnosis, years	23.5 (9.4)	22.5 (8.4)	24.5 (10.4)	24.6 (10.0)	22.3 (8.7)
Sex, n (%)					
Female	224 (89)	120 (90)	104 (87)	120 (92)	104 (85)
Male	29 (11)	14 (10)	15 (13)	11 (8)	18 (15)
Race, n (%)					
Caucasian	210 (83)	111 (83)	99 (83)	103 (79)	107 (88)
Black/African American	24 (9)	11 (8)	13 (11)	16 (12)	8 (7)
Asian/Pacific Islander	8 (3)	6 (4)	2 (2)	4 (3)	4 (3)
Native American	1 (<1)	0	1 (1)	1 (1)	0
Other	7 (3)	3 (2)	4 (3)	4 (3)	3 (2)

CM, chronic migraine; EM, episodic migraine; SD, standard deviation.

Satisfaction With Fremanezumab

- Mean (standard deviation [SD]) overall satisfaction rating for fremanezumab was 6.1 (1.4) out of 7
 - Mean (SD) satisfaction ratings were high across all evaluated aspects of treatment (**Table 3**)
- The majority of patients reported being satisfied overall and with all evaluated aspects of treatment, regardless of migraine classification (CM or EM) or dosing received during the extension study (quarterly or monthly; **Figure 1**)

Table 3. Treatment Satisfaction Scores With Fremanezumab (1 = Extremely Dissatisfied to 7 = Extremely Satisfied)

Satisfaction, mean (SD)	Total (n = 253)	CM (n = 134)	EM (n = 119)	Quarterly (n = 131)	Monthly (n = 122)
Ability to prevent or treat migraine	6.1 (1.4)	5.8 (1.6)	6.5 (0.9)	6.1 (1.3)	6.2 (1.5)
Way the medication relieved symptoms	6.1 (1.4)	5.8 (1.6)	6.4 (1.1)	6.1 (1.3)	6.1 (1.5)
Time it took to start working	5.7 (1.4)	5.5 (1.5)	5.9 (1.3)	5.7 (1.4)	5.7 (1.5)
Ease of medication administration[a]	6.0 (1.4)	5.9 (1.4)	6.1 (1.3)	5.9 (1.5)	6.1 (1.2)
Overall satisfaction with medication	**6.1 (1.4)**	**5.9 (1.6)**	**6.4 (1.1)**	**6.2 (1.3)**	**6.1 (1.5)**

SD, standard deviation; CM, chronic migraine; EM, episodic migraine.
[a]Ratings of 1 = extremely difficult to 7 = extremely easy were used for this question only; extremely dissatisfied to extremely satisfied were used for all other questions and overall satisfaction.

Figure 1. Percentage of patients reporting satisfaction with different aspects of treatment and overall.[a,b]

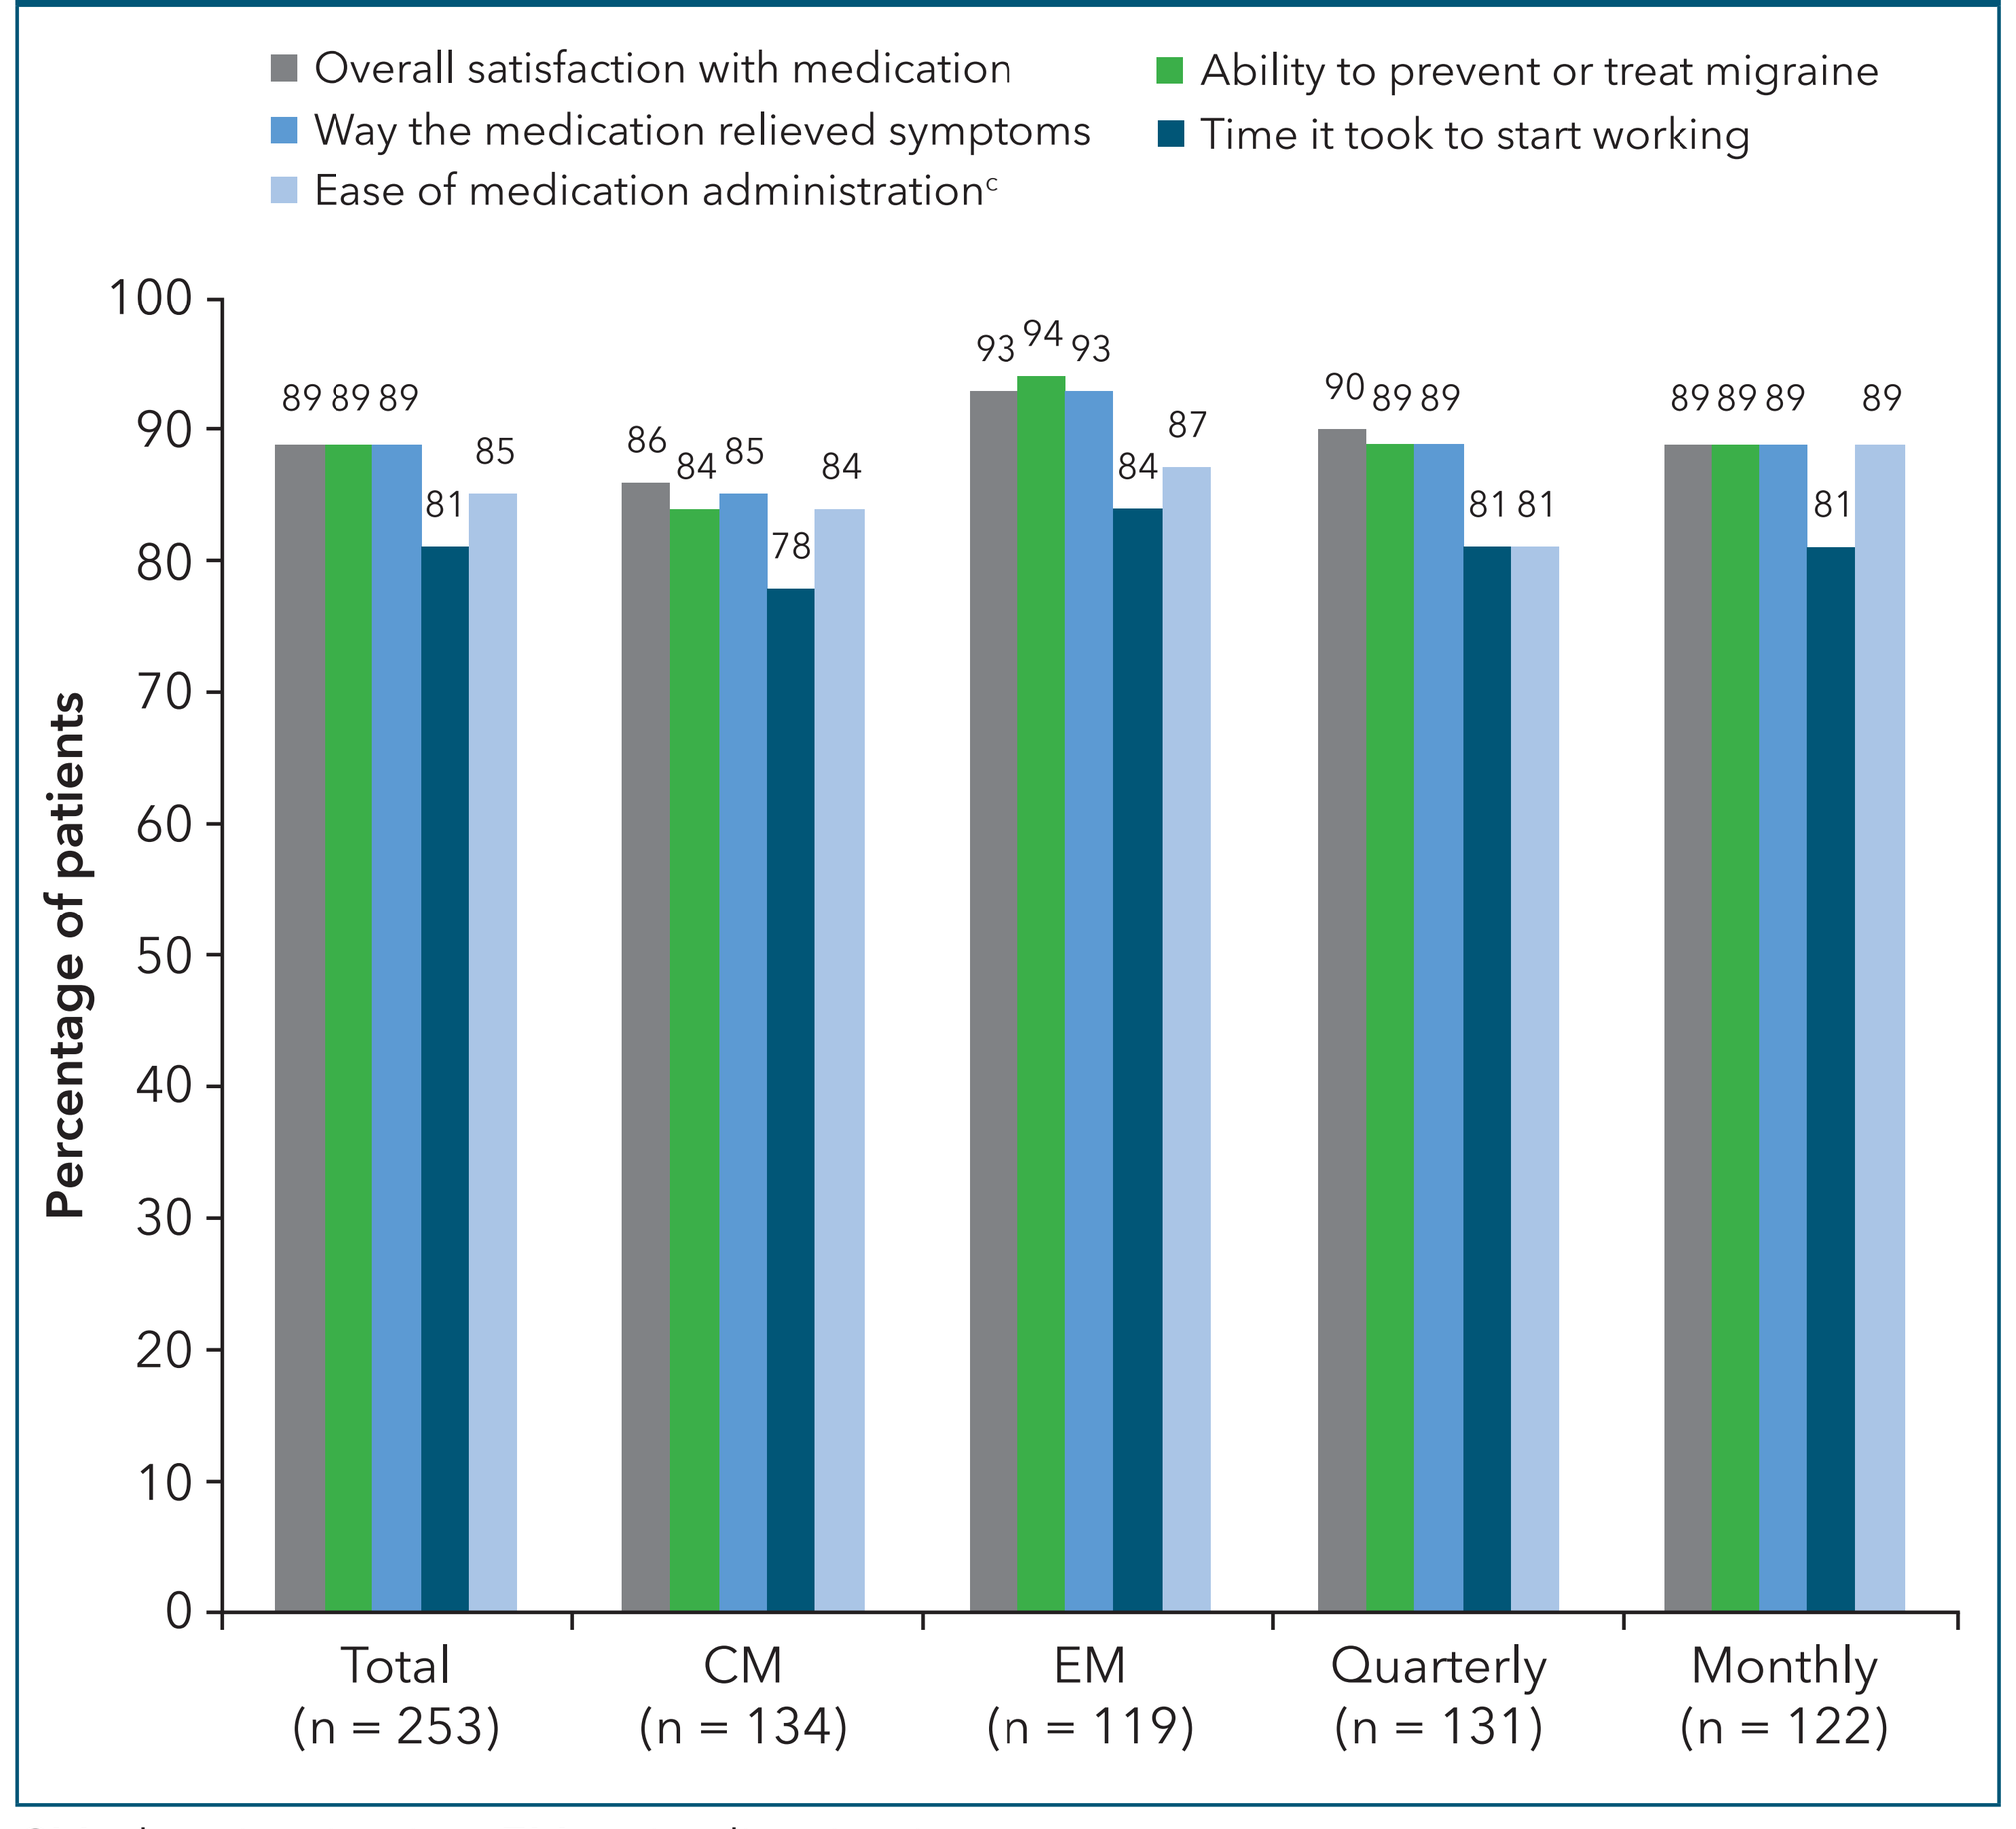
Overall satisfaction with medication
Ability to prevent or treat migraine
Way the medication relieved symptoms
Time it took to start working
Ease of medication administration[c]
Percentage of patients
89 89 89 81 85
86 84 85 78 84
93 94 93 84 87
90 89 89 81 81
89 89 89 81 89
Total (n = 253)
CM (n = 134)
EM (n = 119)
Quarterly (n = 131)
Monthly (n = 122)

CM, chronic migraine; EM, episodic migraine.
[a]Satisfaction overall and with individual dimensions was rated on a Likert scale: 1 = extremely dissatisfied/difficult to 7 = extremely satisfied/easy.
[b]Ratings from 5 to 7 were included here.
[c]Ratings of 1 = extremely difficult to 7 = extremely easy were used for this question only; extremely dissatisfied to extremely satisfied were used for all other questions and overall satisfaction.

- >50% of patients reported satisfaction across every efficacy dimension of fremanezumab (**Figure 2**)

Figure 2. Patient satisfaction with efficacy dimensions of fremanezumab.[a]

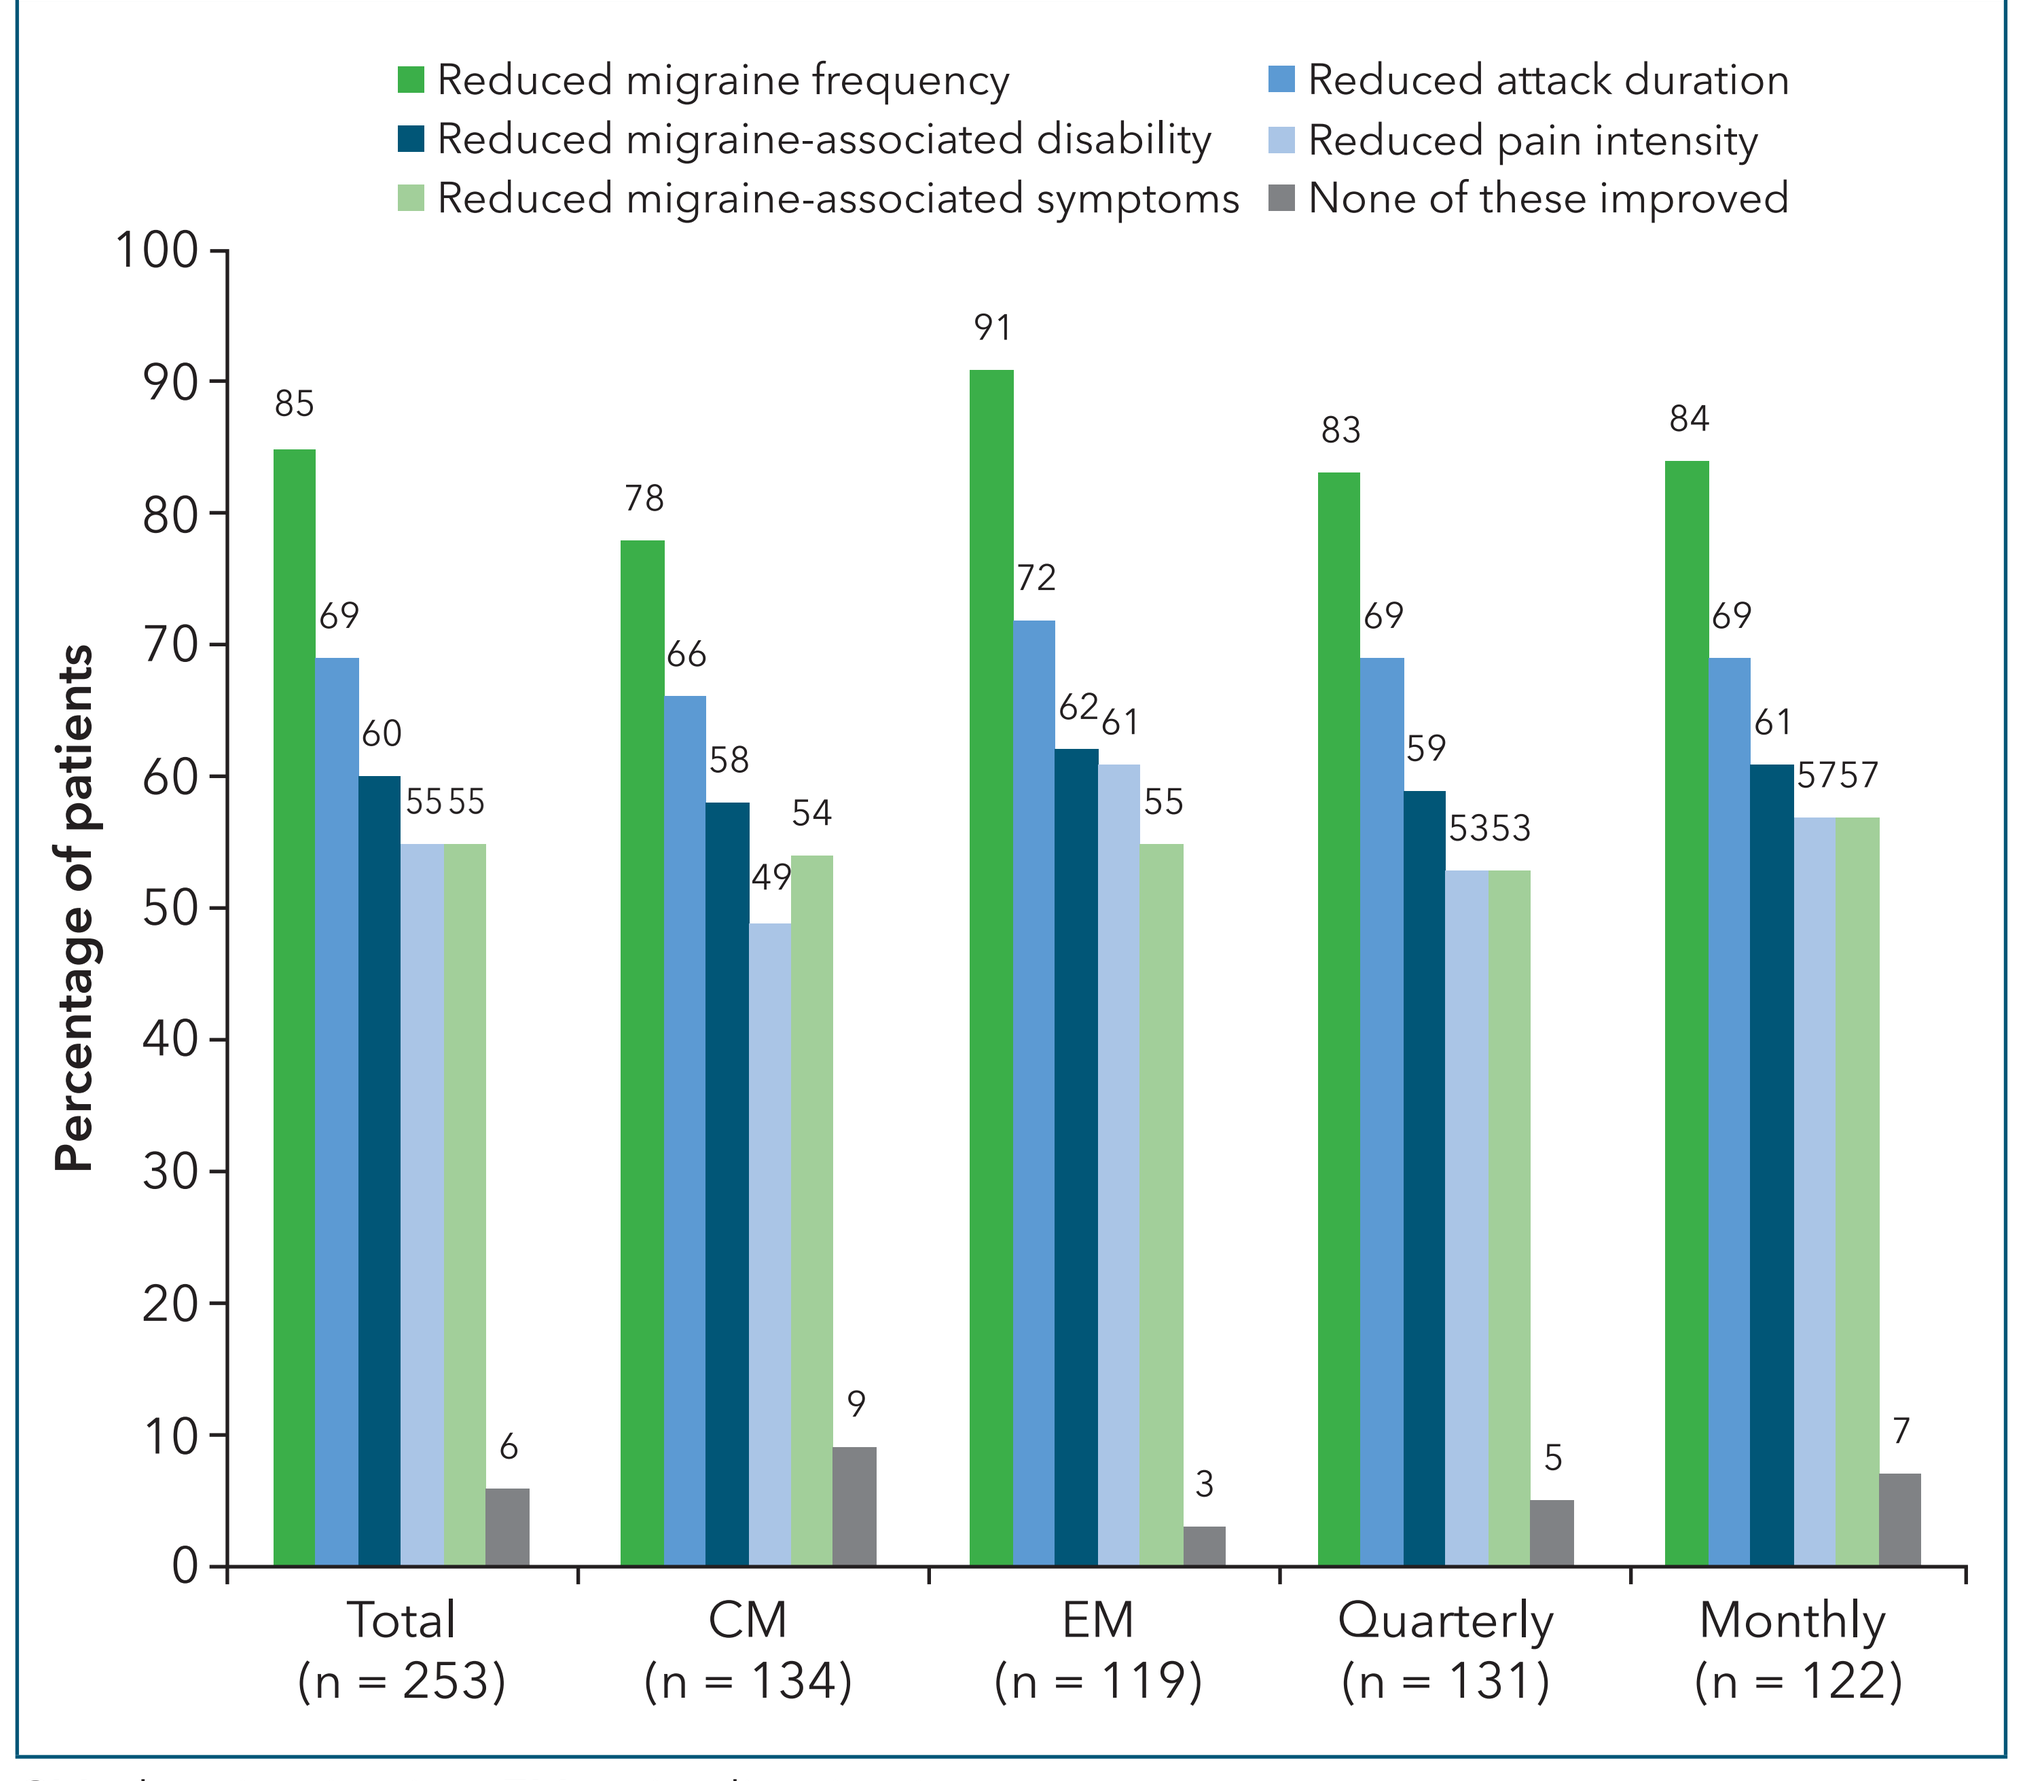
Reduced migraine frequency
Reduced attack duration
Reduced migraine-associated disability
Reduced pain intensity
Reduced migraine-associated symptoms
None of these improved
Percentage of patients
85 69 60 55 55 6
78 66 58 49 54 9
91 72 62 61 55 3
83 69 59 53 53 5
84 69 61 57 57 7
Total (n = 253)
CM (n = 134)
EM (n = 119)
Quarterly (n = 131)
Monthly (n = 122)

CM, chronic migraine; EM, episodic migraine.
[a]Reported in response to the survey question: "Thinking specifically of the experimental medicine's ability to prevent migraine, with which dimension of your migraine relief were you most satisfied? *Select all that apply.*"

References

1. AJOVY® (fremanezumab-vfrm) injection, for subcutaneous use [prescribing information]. Teva Pharmaceuticals USA, Inc.: North Wales, PA; 2019.
2. Bigal ME, et al. *Neurology*. 2016;87(1):41-48.
3. Dodick DW, et al. *JAMA*. 2018;319(19):1999-2008.

Acknowledgments

This study was funded by Teva Branded Pharmaceutical Products R&D, Inc. Medical writing and editorial support were provided by Megan Knagge, PhD, of MedErgy, and were funded by Teva Pharmaceuticals USA, Inc.

Disclosures

R.B. Lipton is the Edwin S. Lowe Professor of Neurology at the Albert Einstein College of Medicine in New York. He receives research support from the NIH: 2PO1 AG003949 (mPI), 5U10 NS077308 (PI), RO1 NS082432 (Investigator), 1RF1 AG057531 (Site PI), RF1 AG054548 (Investigator), 1RO1 AG048642 (Investigator), R56 AG057548 (Investigator), K23 NS09610 (Mentor), K23AG049466 (Mentor), and 1K01AG054700 (Mentor). He also receives support from the Migraine Research Foundation and the National Headache Foundation. He serves on the editorial board of *Neurology*, is a senior advisor to *Headache*, and is the associate editor to *Cephalalgia*. He has reviewed for the NIA and NINDS; holds stock options in eNeura Therapeutics and Biohaven Holdings; and serves as a consultant, serves as an advisory board member, or has received honoraria from the American Academy of Neurology, Alder, Allergan, the American Headache Society, Amgen, Autonomic Technologies, Avanir, Biohaven, Biovision, Boston Scientific, Dr. Reddy's, electroCore, Eli Lilly, eNeura Therapeutics, GlaxoSmithKline, Merck, Pernix, Pfizer, Supernus, Teva Pharmaceuticals, Trigemina, Vector, and Vedanta. He receives royalties from *Wolff's Headache 7th and 8th Edition*, Oxford Press University (2009),Wiley, and Informa. S.K. Gandhi, J.M. Cohen, V. Ramirez-Campos, and A.H. Ahn are employees of Teva Pharmaceuticals. B. Cloud has no conflicts of interest to report. D.C. Buse has received grant support and honoraria from Allergan, Amgen, Avanir, Biohaven, Eli Lilly, Promius, and Teva Pharmaceuticals; and is on the editorial board of *Current Pain and Headache Reports*.

For a copy of this poster, scan the QR code with your Android™ phone, Blackberry®, or iPhone®. No personal information will be collected. This is not associated with any marketing or promotional activity.